Improvement in Atopic Dermatitis Signs and Symptoms With Once-Daily and Proactive Twice-Weekly Roflumilast Cream 0.15% or 0.05%: Results From the 52-Week Phase 3 INTEGUMENT-OLE Trial in Patients Aged ≥2 Years

H. Chih-ho Hong,[1] Eric Simpson,[2] Lawrence F. Eichenfield,[3] Alexandra Golant,[4] Adelaide A. Hebert,[5] Amy Paller,[6] April Armstrong,[7] Linda Stein Gold,[8] Jonathan I. Silverberg,[9] David Krupa,[10] Patrick Burnett,[10] Diane Hanna,[10] Melissa S. Seal,[10] Brett Stephenson[10]

[1]Probity Medical Research and University of British Columbia, Surrey, BC; [2]Oregon Health & Science University, Portland, OR; [3]Rady Children's Hospital-San Diego, University of California, San Diego, CA; [4]Icahn School of Medicine at Mount Sinai, New York, NY; [5]UTHealth McGovern Medical School, Houston, TX; [6]Northwestern University Feinberg School of Medicine, Chicago, IL; [7]David Geffen School of Medicine, University of California Los Angeles, Los Angeles, CA; [8]Henry Ford Health System, Detroit, MI; [9]George Washington University School of Medicine and Health Sciences, Washington, DC; [10]Arcutis Biotherapeutics, Inc., Westlake Village, CA

ABBREVIATIONS

AD, atopic dermatitis; AE, adverse event; BIW, twice weekly; BSA, body surface area affected; EASI, Eczema Area and Severity Index; FAS, full analysis set; K-M, Kaplan-Meier; NE, not evaluable; OLE, open-label extension; PDE4, phosphodiesterase 4; PED, pediatric; ROF, roflumilast; QD, once daily; SAE, serious AE; TCS, topical corticosteroids; TEAE, treatment-emergent AE; US, United States; vIGA-AD, Validated Investigator Global Assessment for AD; WI-NRS, Worst Itch-Numeric Rating Scale; y, years.

REFERENCES

1. Silverberg JI, et al. *Ann Allergy Asthma Immunol*. 2021;126:417–428. 2. Napolitano M, et al. *Dermatologic Ther*. 2022;35:e15901. 3. Sathishukumar D, et al. *Curr Med Issues*. 2020;18:317–325. 4. AAAAI/ACAAI JTF Atopic Dermatitis Guideline Panel. *Ann Allergy Asthma Immunol*. 2023;132:274–312. 5. Burshtein J, et al. *Dermatol Online J*. 2025;31(1). doi:10.5070/D331164978. 6. Draelos ZD, et al. *J Drugs Dermatol*. 2024;23:834–840. 7. ZORYVE® (roflumilast) cream 0.15%. Prescribing information. Arcutis Biotherapeutics, Inc. July 2024. 8. Eichenfield LF, et al. *Pediatr Dermatol*. 2025;42:296–304. 9. Simpson EL, et al. *Dermatitis*. Available online January 10, 2025; doi:10.1089/derm.2024.0418. 10. Simpson EL, et al. *JAMA Dermatol*. 2024;160:1161–1170. 11. Eichenfield LF, et al. Poster presented at: 2025 RAD Conference. June 6–7, 2025; Nashville, TN.

ACKNOWLEDGMENTS

Thank you to the investigators and their staff for their participation in the trials. We are grateful to the study participants and their families for their time and commitment. Writing support was provided by Kelly M. Fahrbach, PhD, CMPP, and Andrea Michels, of Ashfield MedComms, an Inizio company, and was funded by Arcutis Biotherapeutics, Inc.

DISCLOSURES

This study was funded by Arcutis Biotherapeutics, Inc. HCH, ES, LFE, AG, AAH, AP, AA, LSG, and JIS are investigators and/or consultants for and received grants/research funding and/or honoraria from Arcutis Biotherapeutics, Inc. DK, PB, DH, MSS, and BS are employees of Arcutis Biotherapeutics, Inc. Additional disclosures provided upon request.

Presented at the 2025 Revolutionizing Atopic Dermatitis Conference
June 6–7, 2025; Nashville, TN

INTRODUCTION

- AD is a chronic inflammatory skin disease that is often first diagnosed during childhood and has a prevalence of ~10% among those aged 6 months to <6 years in the US[1,2]
 - Signs and symptoms of AD can result in comorbidities and negatively impact quality of life of the whole family[3]
- Despite advances in the development of topical treatments that are not steroids, TCS are commonly used to treat AD; however, there are several limitations to their use, even more so in younger patients[4,5]
 - Because of increased risk of cutaneous and systemic AEs, TCS are not approved for long-term use and higher potency TCS are not recommended for thin-skinned areas (eg, face, younger skin) where there is greater systemic absorption[3–5]
 - Children with less developed skin barriers and greater BSA to weight ratios are at even greater risk for AEs
- Roflumilast, a potent PDE4 inhibitor, has been formulated as a topical cream or foam that does not include propylene glycol, formaldehyde, fragrances, or potential cutaneous irritants[6]
 - Roflumilast cream 0.15% is approved for treatment of mild-to-moderate AD in patients aged ≥6 years[7]
 - Efficacy and safety of once-daily roflumilast cream 0.15% and 0.05% in patients with AD aged ≥6 years and 2–5 years, respectively, were demonstrated in the 4-week, phase 3 INTEGUMENT-1/2 (NCT04662487/NCT04773600)[8] and INTEGUMENT-PED (NCT04845620)[9] trials
- Use of roflumilast cream 0.15% for up to 56 weeks in patients aged ≥2 years with AD was investigated in the INTEGUMENT-OLE (NCT04804605) study
 - Primary safety and efficacy outcomes in patients aged ≥6 years from the OLE study are reported by Eichenfield, et al[10]
- Long-term outcomes, including changes in BSA from patients aged ≥6 years and 2–5 years from INTEGUMENT-OLE, are reported here

METHODS

Study design

- INTEGUMENT-OLE was a 52-week, phase 3, multicenter, OLE trial in patients aged ≥2 years with AD
 - Patients who completed 4 weeks in one of the parent studies (INTEGUMENT-1/2, ≥6 years or INTEGUMENT-PED, 2–5 years) with no safety concerns were allowed to enroll in the INTEGUMENT-OLE trial and apply once-daily roflumilast cream 0.15% or 0.05%, respectively
 - Patients who 'aged up' from 5 to 6 years during the study were switched to roflumilast cream 0.15% at their first scheduled visit after their birthday
- Patients could switch to BIW application any time after week 4, if they achieved vIGA-AD 0
 - BIW treatment was maintained if signs and symptoms were adequately controlled and vIGA-AD remained clear or almost clear (0/1)

Assessments in this analysis

- vIGA-AD 0/1
- vIGA-AD success, defined as vIGA-AD 0/1 plus a ≥2-point improvement from parent study baseline
- WI-NRS success, defined as ≥4-point improvement in patients with parent study baseline WI-NRS ≥4
- BSA over time
- 'Disease control', defined as duration of vIGA-AD 0/1 and adequate control of signs and symptoms with BIW application

Study Design

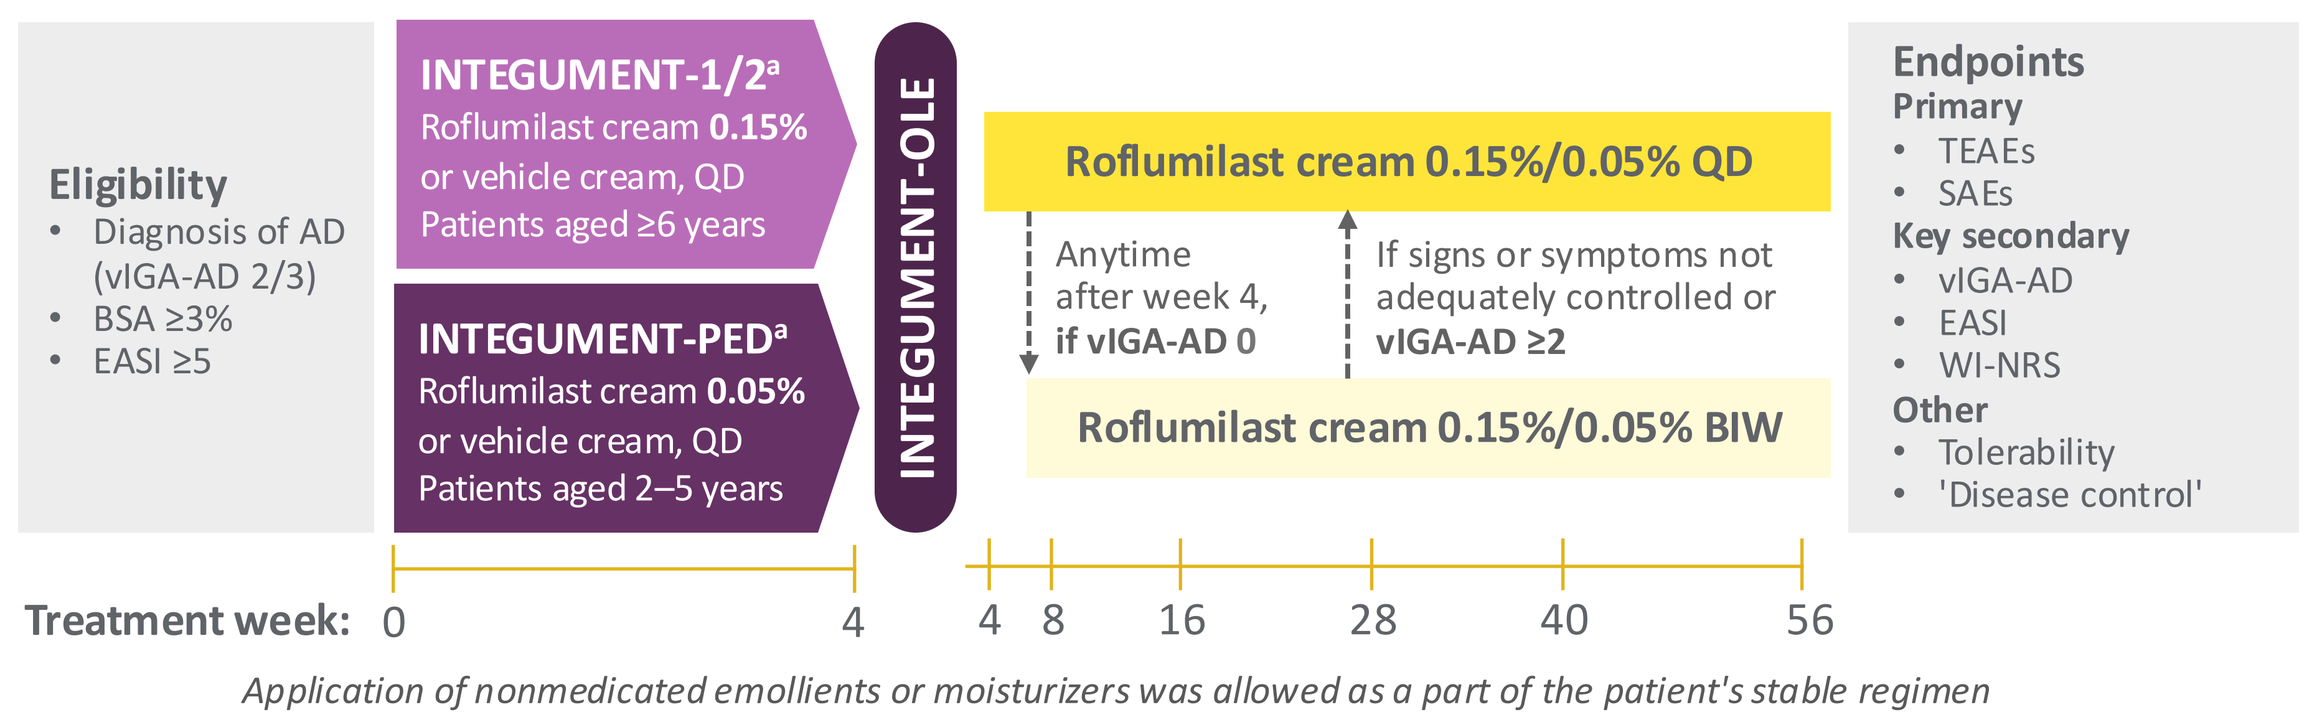
Eligibility
• Diagnosis of AD (vIGA-AD 2/3)
• BSA ≥3%
• EASI ≥5
INTEGUMENT-1/2a
Roflumilast cream 0.15% or vehicle cream, QD
Patients aged ≥6 years
INTEGUMENT-PEDa
Roflumilast cream 0.05% or vehicle cream, QD
Patients aged 2–5 years
INTEGUMENT-OLE
Roflumilast cream 0.15%/0.05% QD
Anytime after week 4, if vIGA-AD 0
If signs or symptoms not adequately controlled or vIGA-AD ≥2
Roflumilast cream 0.15%/0.05% BIW
Endpoints
Primary
• TEAEs
• SAEs
Key secondary
• vIGA-AD
• EASI
• WI-NRS
Other
• Tolerability
• 'Disease control'
Treatment week: 0 4 4 8 16 28 40 56
Application of nonmedicated emollients or moisturizers was allowed as a part of the patient's stable regimen

[a]After OLE study enrollment commenced, the protocol was amended to allow patients (aged 2–5 years) who completed INTEGUMENT-PED to enroll.

RESULTS

- There were 658 patients from INTEGUMENT-1/2 and 562 patients from INTEGUMENT-PED who entered the INTEGUMENT-OLE study
 - Patient demographics and baseline clinical characteristics were balanced among those who continued treatment with roflumilast cream and those who received vehicle in the parent study and switched to roflumilast cream in the OLE study[9,10]
- Roflumilast cream 0.15%/0.05% was well tolerated, as previously reported[8–11]
 - Treatment-related AEs were reported for 4.7%[9] and 2.5% of patients who entered from the INTEGUMENT-1/2 and INTEGUMENT-PED trials, respectively
 - Application-site pain was reported as an AE for 0.5% of patients from INTEGUMENT-1/2[9] and 0.7% of patients from INTEGUMENT-PED
- Improvements in AD observed in the parent trials were maintained or continued to improve throughout the end of the OLE study
 - vIGA-AD 0/1 was achieved by 55.7% and 63.1% of patients from INTEGUMENT-1/2 and INTEGUMENT-PED, respectively
 - WI-NRS 0/1 was achieved by 47.1% and 40.7% of patients, respectively
- Mean BSA decreased from baseline of each parent study through the end of the OLE
 - Patients from INTEGUMENT-1/2: 14.8% at baseline to 3.7%
 - Patients from INTEGUMENT-PED: 22.3% at baseline to 4.9%
- Patients who achieved vIGA-AD 0 and switched to BIW application maintained a median duration of 'disease control' (K-M estimates) of ≥238 days (34 weeks)

Durable Improvements in AD Outcomes[a]

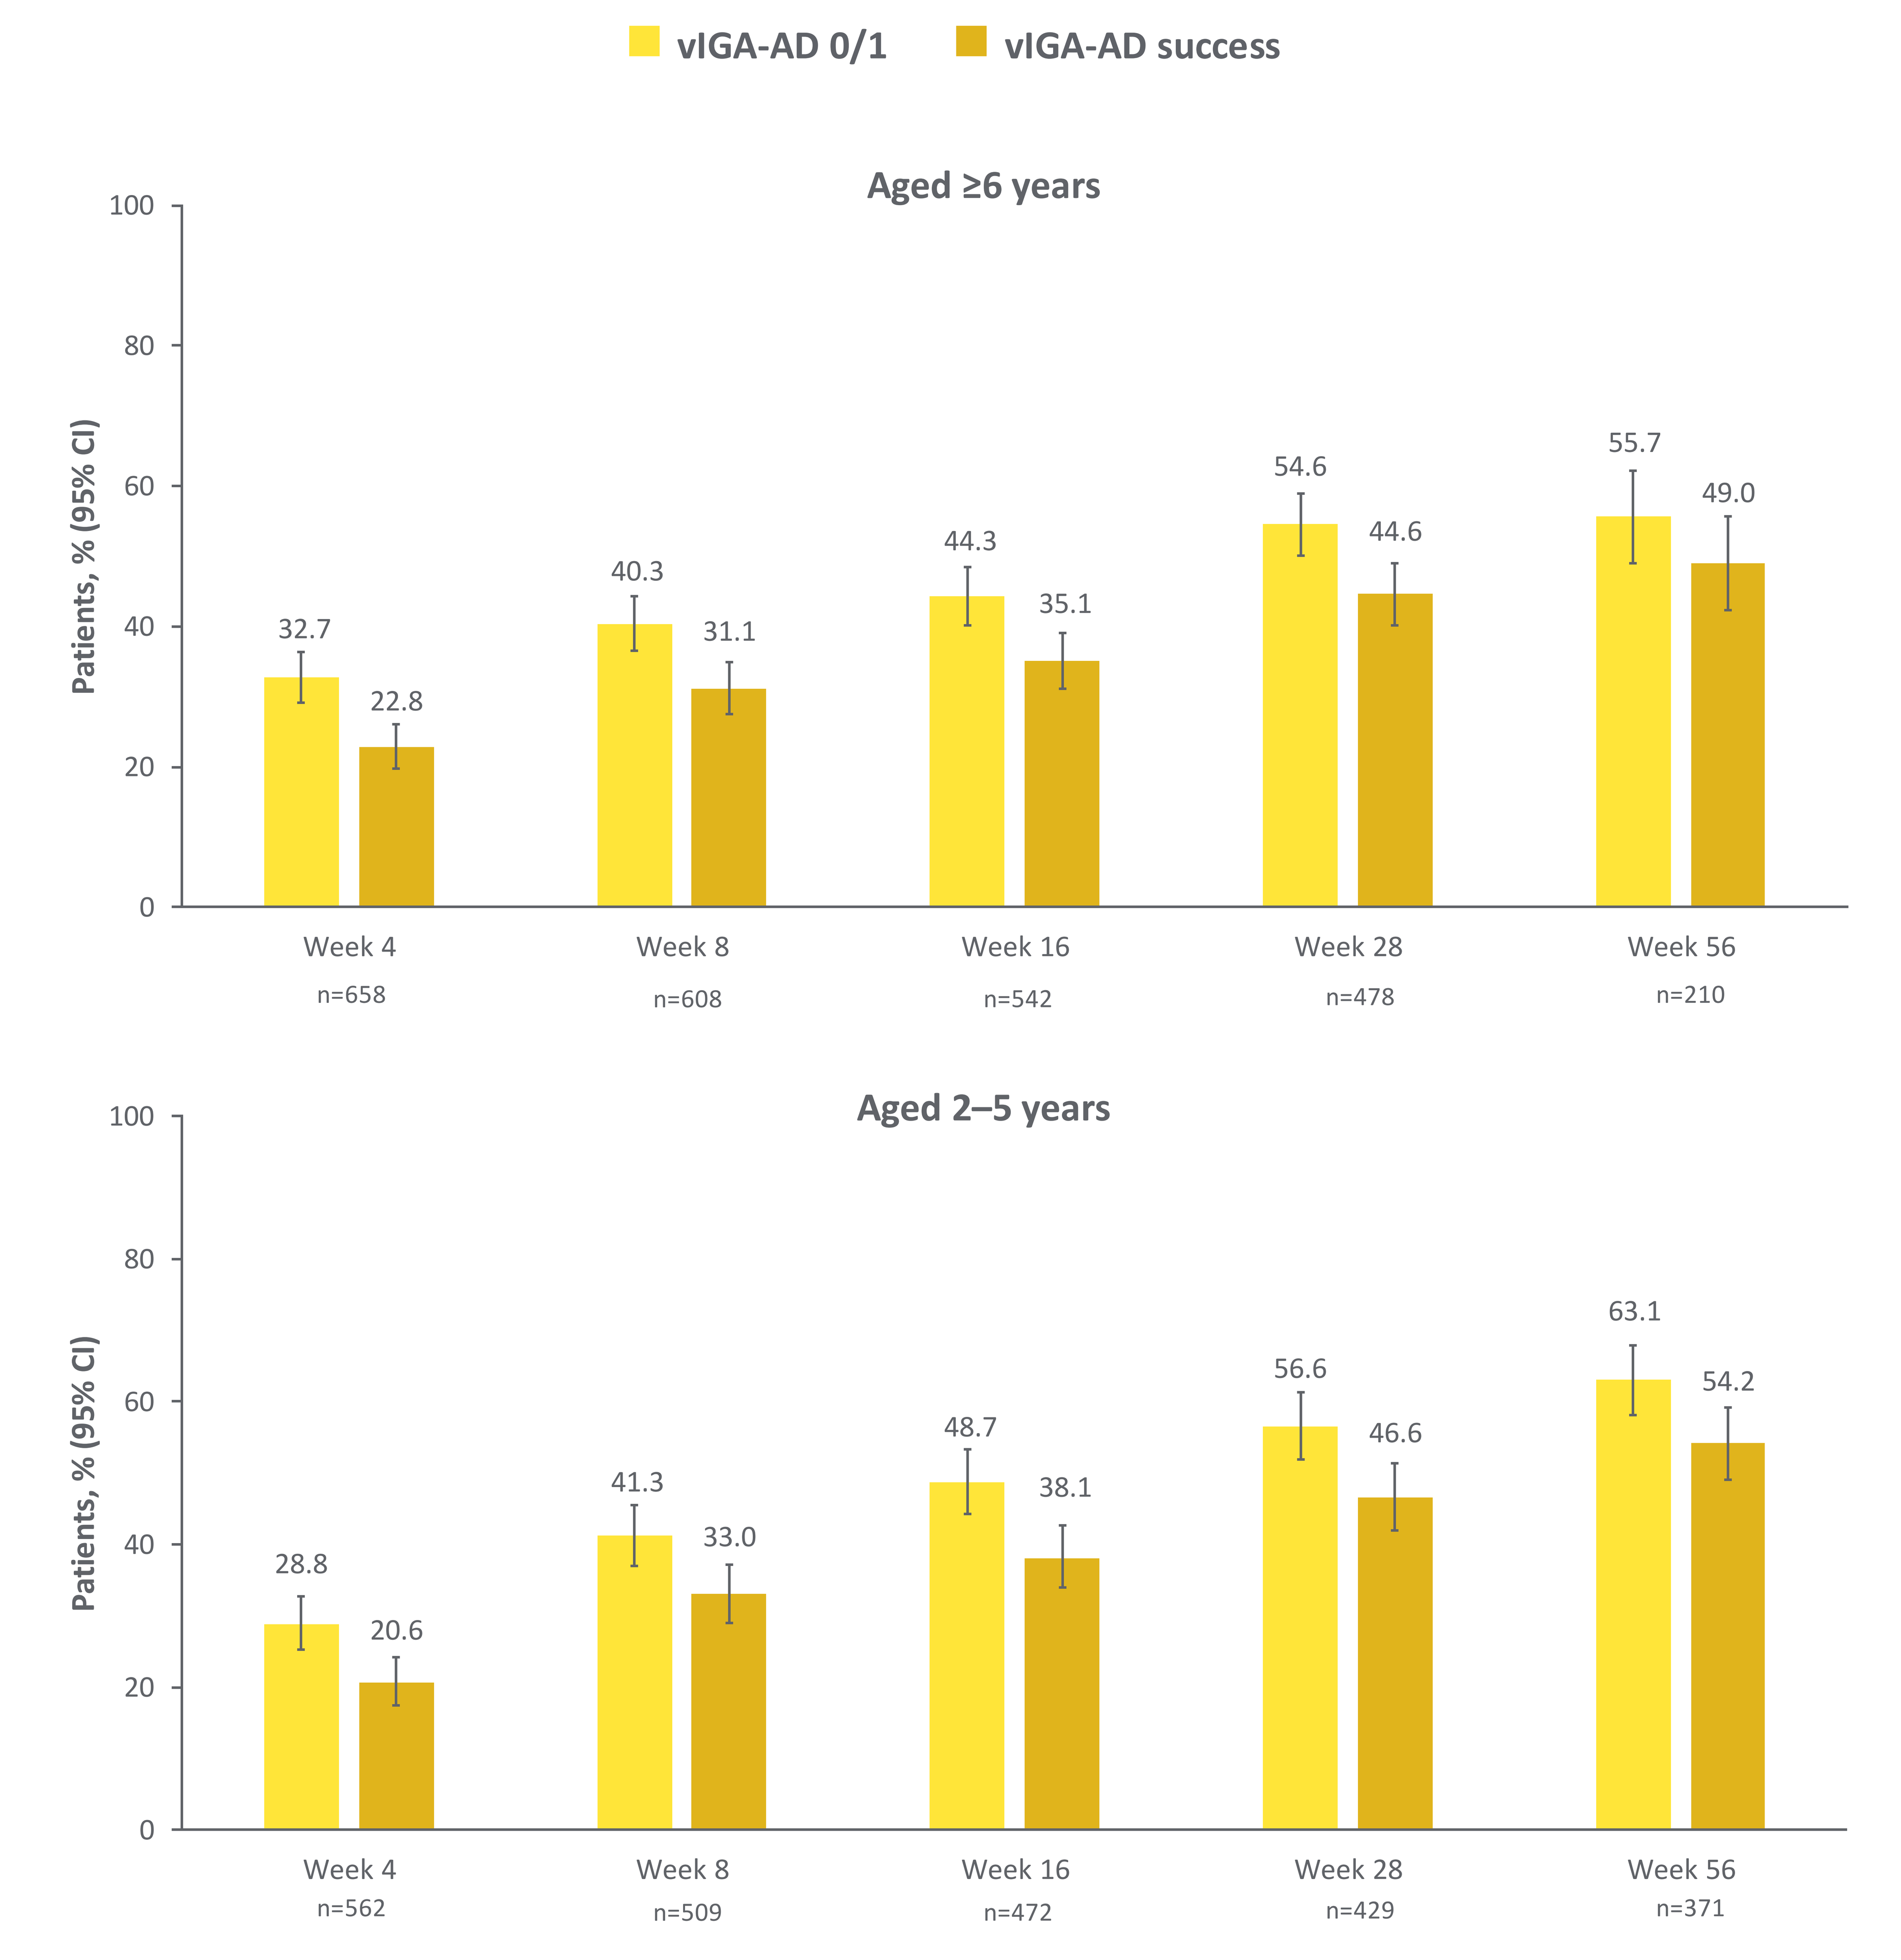
vIGA-AD 0/1
vIGA-AD success
Aged ≥6 years
Patients, % (95% CI)
100
80
60
40
20
0
32.7
22.8
40.3
31.1
44.3
35.1
54.6
44.6
55.7
49.0
Week 4
n=658
Week 8
n=608
Week 16
n=542
Week 28
n=478
Week 56
n=210
Aged 2–5 years
28.8
20.6
41.3
33.0
48.7
38.1
56.6
46.6
63.1
54.2
Week 4
n=562
Week 8
n=509
Week 16
n=472
Week 28
n=429
Week 56
n=371

FAS; observed cases. [a]Over time by treatment week.

Durable Improvement in Itch Symptoms[a]

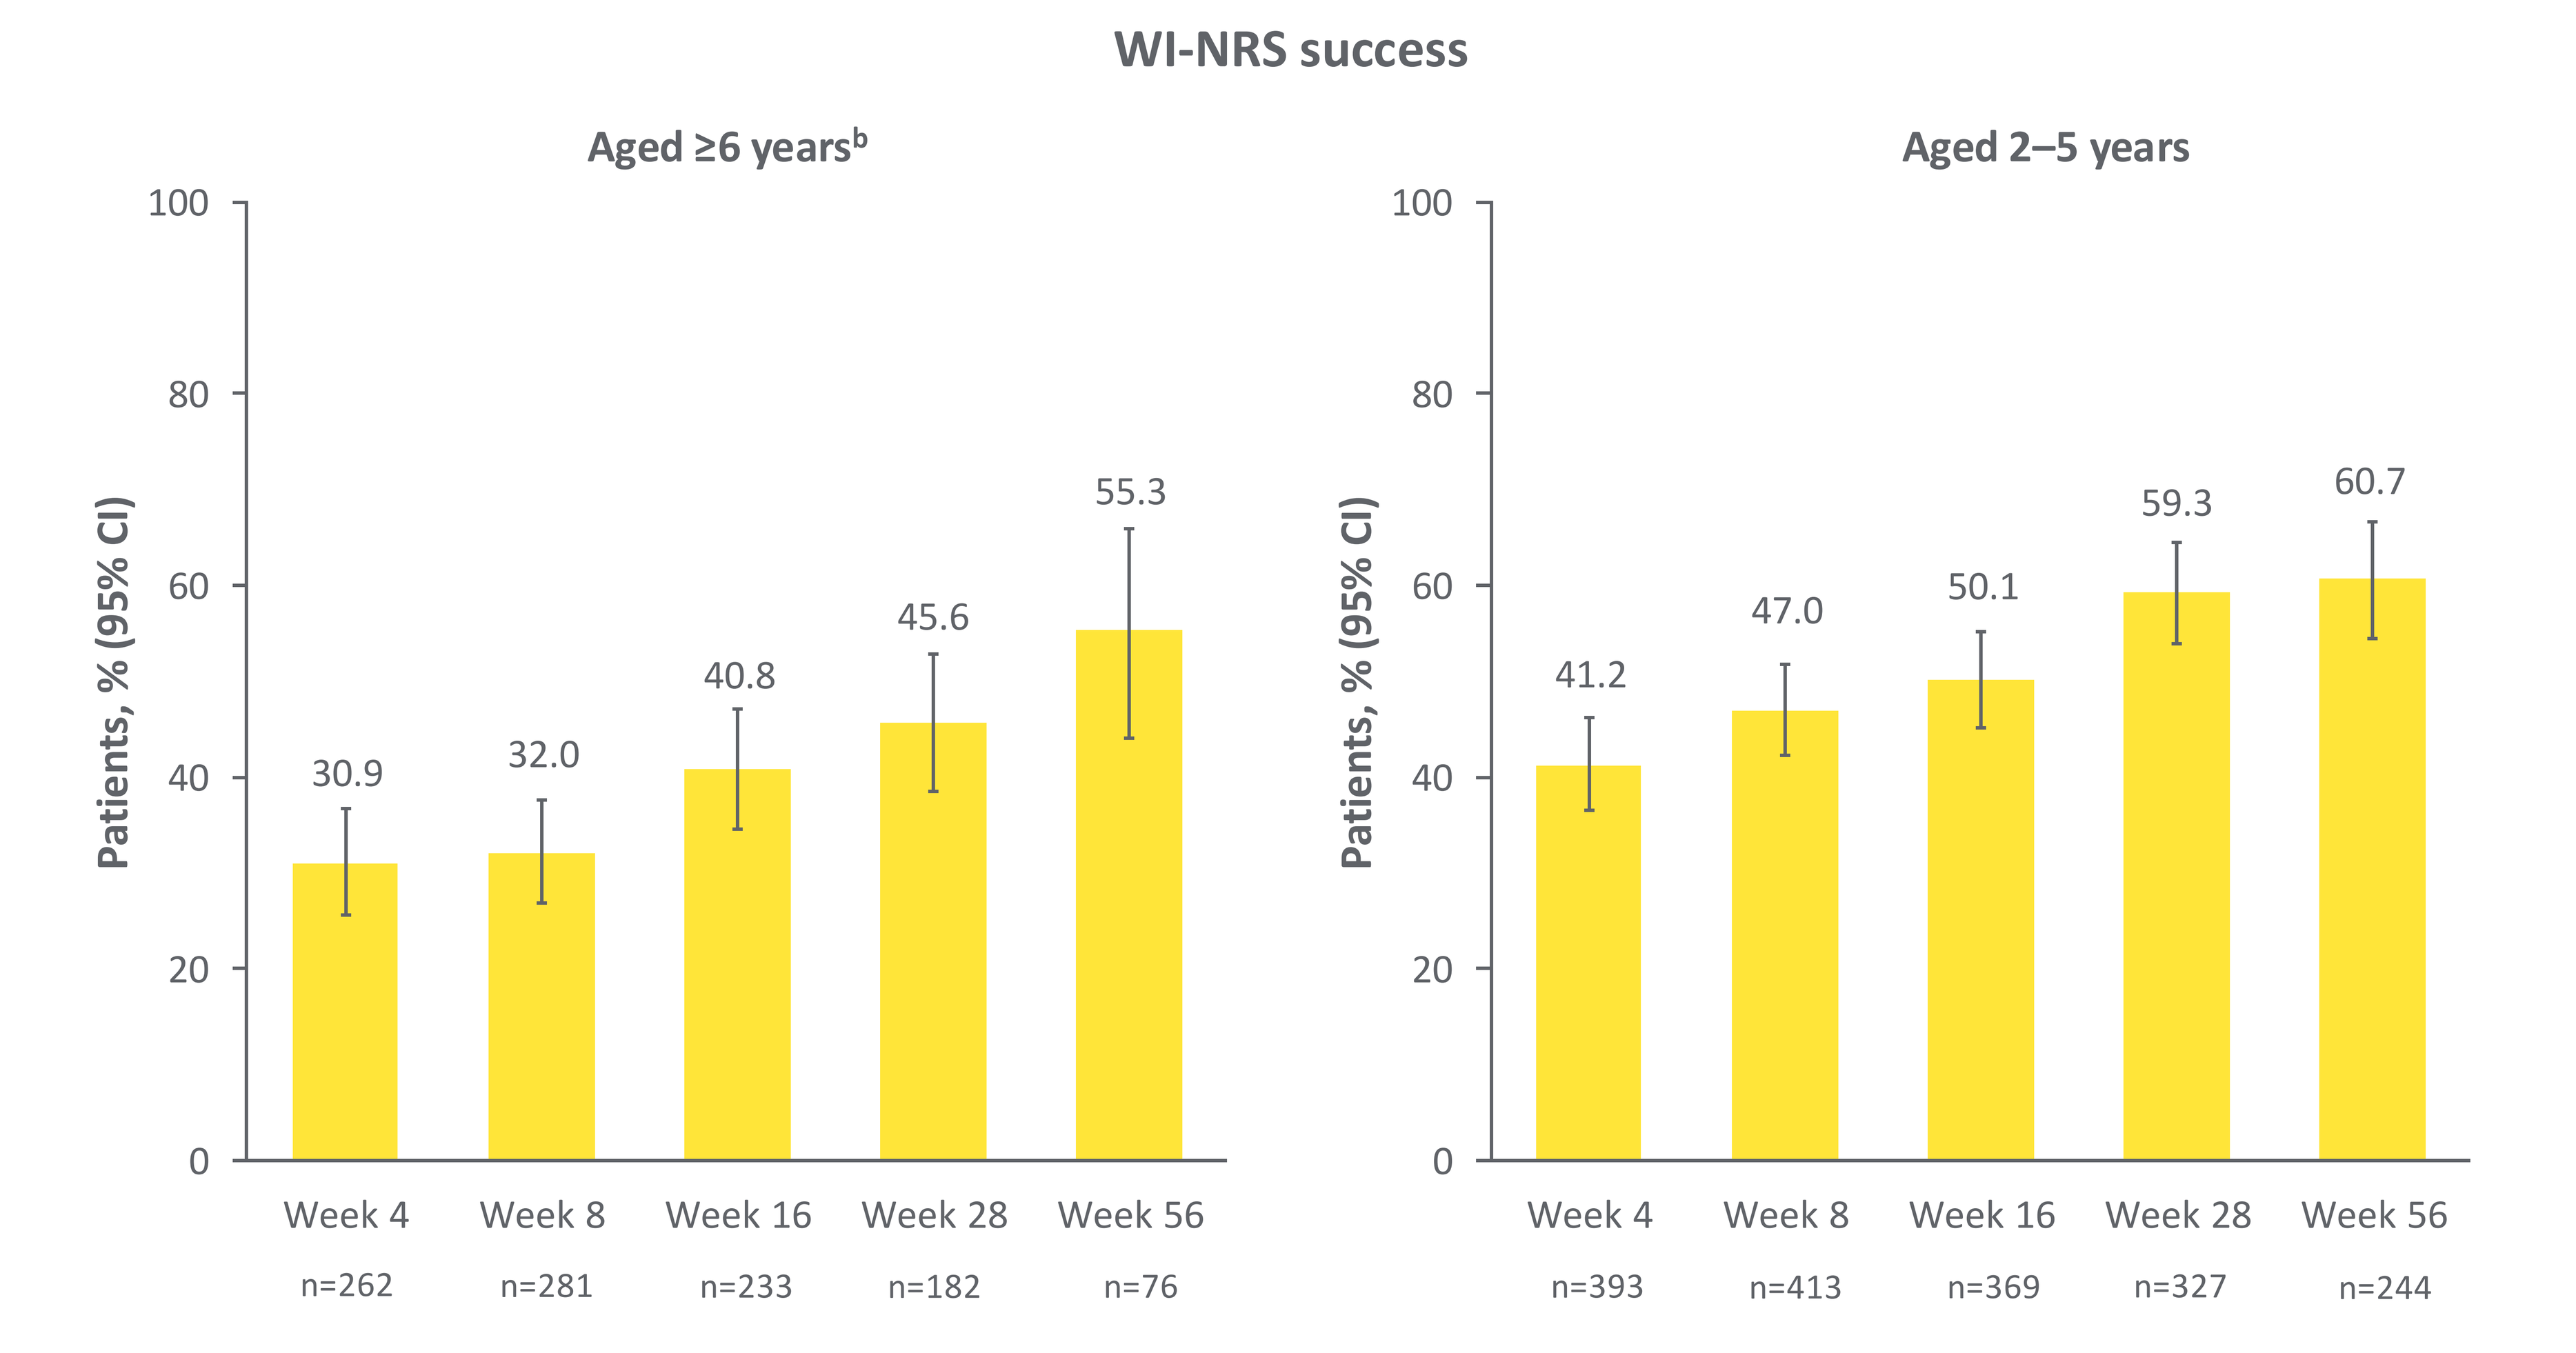
WI-NRS success
Aged ≥6 yearsb
Patients, % (95% CI)
100
80
60
40
20
0
30.9
32.0
40.8
45.6
55.3
Week 4
n=262
Week 8
n=281
Week 16
n=233
Week 28
n=182
Week 56
n=76
Aged 2–5 years
41.2
47.0
50.1
59.3
60.7
Week 4
n=393
Week 8
n=413
Week 16
n=369
Week 28
n=327
Week 56
n=244

FAS; observed cases. [a]Over time by treatment week. [b]In patients aged ≥12 years.

BSA Over Time

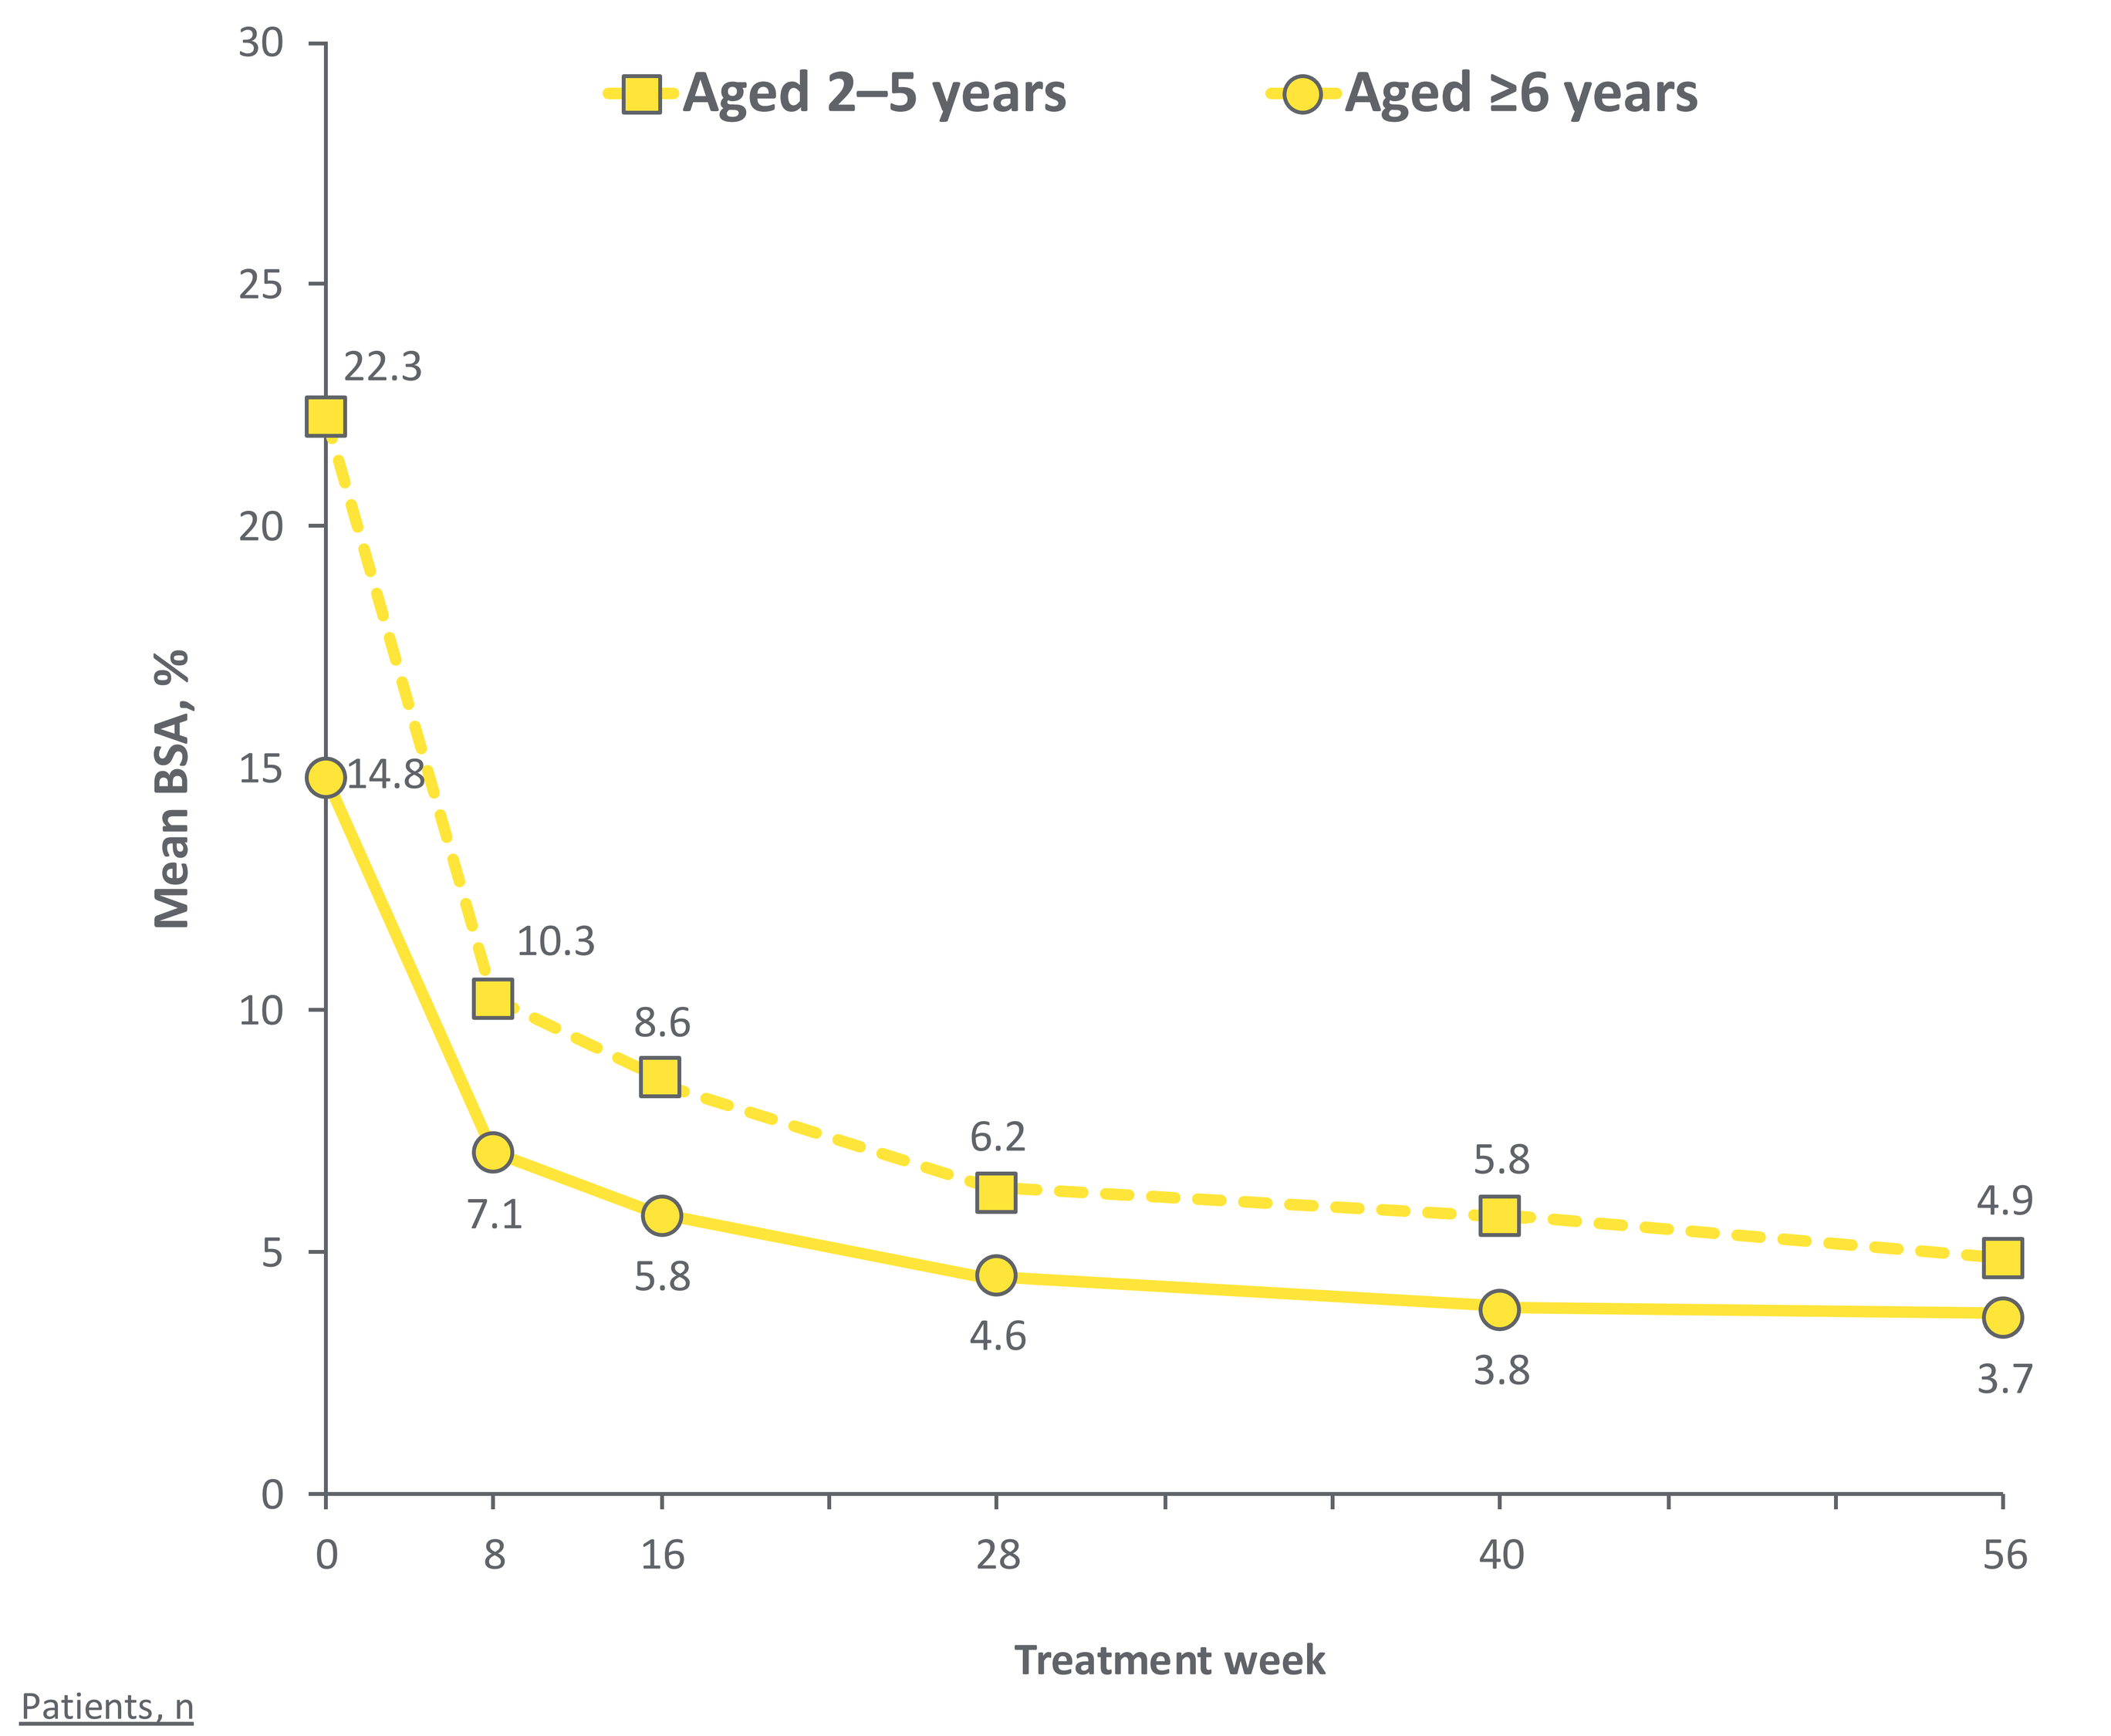
Aged 2–5 years
Aged ≥6 years
Mean BSA, %
30
25
20
15
10
5
0
22.3
14.8
10.3
7.1
8.6
5.8
6.2
4.6
5.8
3.8
4.9
3.7
0
8
16
28
40
56
Treatment week

Patients, n	0	8	16	28	40	56
Aged 2–5 years	562	535	495	440	396	374
Aged ≥6 years	658	629	568	486	228	210

FAS; observed cases.

'Disease Control' With Proactive BIW Application

	Achieved vIGA-AD 0 and switched to proactive BIW application	Median duration of 'disease control'[c] with BIW application
Aged ≥6 years[a]	130 of 658 patients (19.8%)	281 days (95 CI%, 147–NE)
Aged 2–5 years[b]	170 of 562 patients (30.2%)	238 days (95 CI%, 146–331)

[a]Patients entering the OLE from INTEGUMENT-1/2. [b]Patients entering the OLE from INTEGUMENT-PED. [c]K-M median.

CONCLUSIONS

- Roflumilast cream 0.15% and 0.05% decreased signs and symptoms of AD and maintained improvements through up to 56 weeks of treatment in patients aged ≥2 years
 - In addition to durable improvements in vIGA-AD and itch symptoms (WI-NRS), BSA decreased after 4 weeks of treatment in parent trials, and was maintained or improved further over 52 weeks of treatment in the OLE
- Patients who achieved clear skin (vIGA-AD 0) with QD application and transitioned to proactive BIW application were able to maintain 'disease control' (ie, vIGA-AD 0/1 and adequate control of signs and symptoms)
- Overall, these results indicate that roflumilast cream is well tolerated and an appropriate alternative to topical therapies that do not offer long-term disease control (eg, TCS or calcineurin inhibitors) for patients with AD