Long-Term Safety and Maintenance of Efficacy With Once-Daily Roflumilast Cream 0.05% in 2–5-year-olds With Atopic Dermatitis: Data From a 52-Week Phase 3 Trial (INTEGUMENT-OLE)

Lawrence F. Eichenfield,[1] Mercedes E. Gonzalez,[2] Mark Boguniewicz,[3] Tracy Funk,[4] Adelaide A. Hebert,[5] Vimal H. Prajapati,[6] Chesahna Kindred,[7] Rocco Serrao,[8] Lisa Swanson,[9] David Krupa,[10] Diane Hanna,[10] Melissa S. Seal,[10] Patrick Burnett,[10] David R. Berk[10]

[1]Rady Children's Hospital-San Diego and University of California San Diego, San Diego, CA; [2]Pediatric Skin Research, LLC, Coral Gables, FL; [3]National Jewish Health, Denver, CO and University of Colorado School of Medicine, Aurora, CO; [4]Oregon Health & Science University, Portland, OR; [5]UTHealth McGovern Medical School, Houston, TX; [6]Dermatology Research Institute, Probity Medical Research, Skin Health & Wellness Centre, and University of Calgary, Calgary, AB; [7]Kindred Hair and Skin Center, Marriottsville, MD; [8]DOCS Dermatology, Mason, OH; [9]Ada West Dermatology, Meridian, ID; [10]Arcutis Biotherapeutics, Inc., Westlake Village, CA

ABBREVIATIONS

AD, atopic dermatitis; AE, adverse event; BIW, twice weekly; BSA, body surface area affected; EASI, Eczema Area and Severity Index; FAS, full analysis set; K-M, Kaplan-Meier; OLE, open-label extension; PDE4, phosphodiesterase 4; PED, pediatric; QD, once daily; ROF, roflumilast; SAE, serious AE; TCS, topical corticosteroids; TEAE, treatment-emergent AE; vIGA-AD, Validated Investigator Global Assessment for AD; WI-NRS, Worst Itch-Numeric Rating Scale.

REFERENCES

1. Silverberg JI, et al. *Ann Allergy Asthma Immunol.* 2021;126:417–428. 2. Abuabara K, Margolis DJ. *J Allergy Clin Immunol.* 2013;132:1139–1140. 3. AAAAI/ACAAI JTF Atopic Dermatitis Guideline Panel. *Ann Allergy Asthma Immunol.* 2023;132:274–312. 4. Burshtein J, et al. *Dermatol Online J.* 2025;31(1). doi:10.5070/D331164978. 5. Draelos ZD, et al. *J Drugs Dermatol.* 2024;23:834–840. 6. ZORYVE® (roflumilast) cream 0.15%. Prescribing information. Arcutis Biotherapeutics, Inc. July 2024. 7. Eichenfield LF, et al. *Pediatr Dermatol.* 2025;42:296–304. 8. Simpson EL, et al. *Dermatitis.* Available online January 10, 2025; doi:10.1089/derm.2024.0418.

ACKNOWLEDGMENTS

Thank you to the investigators and their staff for their participation in the trials. We are grateful to the study participants and their families for their time and commitment. Writing support was provided by Kelly M. Fahrbach, PhD, CMPP, and Andrea Michels, of Ashfield MedComms, an Inizio company, and was funded by Arcutis Biotherapeutics, Inc.

DISCLOSURES

This study was funded by Arcutis Biotherapeutics, Inc. LFE, MEG, MB, TF, AAH, VHP, CK, RS, and LS are investigators and/or consultants for and received grants/research funding and/or honoraria from Arcutis Biotherapeutics, Inc.; DK, DH, MSS, PB, and DRB are employees of Arcutis Biotherapeutics, Inc. Additional disclosures provided upon request.

Presented at the 2025 Revolutionizing Atopic Dermatitis Conference
June 6–7, 2025; Nashville, TN

INTRODUCTION

- AD is a chronic condition, often initially diagnosed during childhood and potentially requiring life-long treatment[1,2]
- TCS are commonly used to treat AD, despite limitations to their use[3,4]
 - Because of increased risk of cutaneous and systemic AEs, TCS are not approved for long-term use and higher potency TCS are not recommended for thin-skinned areas with greater systemic absorption
- Roflumilast is a potent PDE4 inhibitor that has been formulated as a topical cream or foam that does not include propylene glycol, formaldehyde, fragrances, or potential cutaneous irritants[5]
 - Roflumilast cream 0.15% is approved for treatment of mild-to-moderate AD in patients aged ≥6 years[6]
- Efficacy and safety of once-daily roflumilast cream 0.05% in children aged 2–5 years with AD were demonstrated in the 4-week, phase 3 INTEGUMENT-PED (NCT04845620) trial[7]
- Roflumilast cream 0.15%/0.05% treatment of patients aged ≥2 years with AD for up to 56 weeks was investigated in an OLE trial (INTEGUMENT-OLE/NCT04804605)
 - Patients aged ≥6 years who completed one of the 4-week parent trials (INTEGUMENT-1 or INTEGUMENT-2) and continued to apply roflumilast cream 0.15% in the INTEGUMENT-OLE trial maintained the improvement in signs and symptoms of AD that were observed during the parent trials[8]
 - The outcomes for patients aged 2–5 years who enrolled in INTEGUMENT-OLE after completing INTEGUMENT-PED are reported here

METHODS

Study design

- INTEGUMENT-OLE was a 52-week, phase 3, multicenter, OLE trial in adults and children aged ≥2 years with AD
 - Patients who completed a 4-week parent trial (INTEGUMENT-1, INTEGUMENT-2, or INTEGUMENT-PED) with no safety concerns were allowed to enroll in the INTEGUMENT-OLE trial and apply once-daily roflumilast cream 0.05% for up to 52 weeks
 - Patients who 'aged up' from 5 to 6 years during the study were to be switched to roflumilast cream 0.15% at their first scheduled visit after their birthday
- Any time after week 4, patients were to be switched to BIW application when they achieved vIGA-AD of clear (0)
 - BIW application was to be maintained as long as symptoms were adequately controlled and vIGA-AD remained clear or almost clear (0/1)

Endpoints and assessments

- AEs and SAEs (primary endpoint)
- vIGA-AD 0/1
- vIGA-AD success, defined as vIGA-AD 0/1 plus a 2-point improvement from baseline
- WI-NRS success, defined as ≥4-point reduction in WI-NRS score from baseline for patients with a baseline score ≥4
- EASI-75, defined as ≥75% improvement in EASI from baseline
- Investigator- and patient-rated application-site tolerability
- 'Disease control', defined as duration of vIGA-AD 0/1 and adequate control of symptoms on BIW application

Study Design

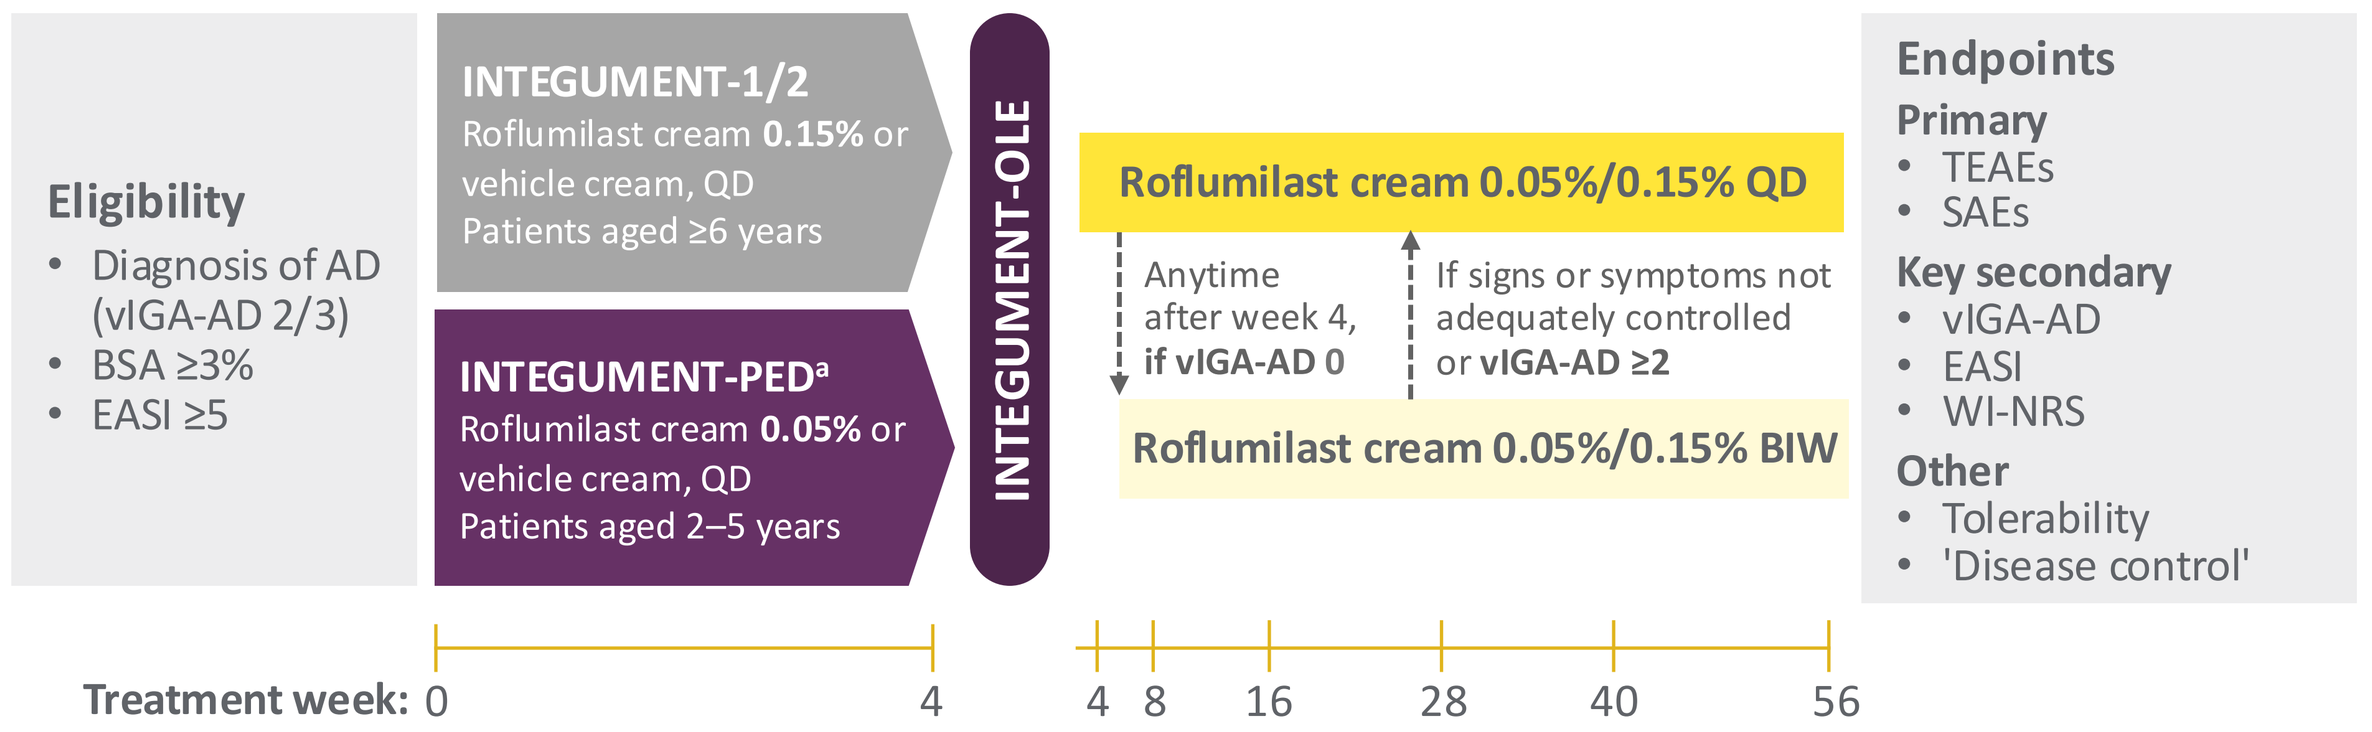

Eligibility
• Diagnosis of AD (vIGA-AD 2/3)
• BSA ≥3%
• EASI ≥5
INTEGUMENT-1/2
Roflumilast cream 0.15% or vehicle cream, QD
Patients aged ≥6 years
INTEGUMENT-PED[a]
Roflumilast cream 0.05% or vehicle cream, QD
Patients aged 2–5 years
INTEGUMENT-OLE
Roflumilast cream 0.05%/0.15% QD
Anytime after week 4, if vIGA-AD 0
If signs or symptoms not adequately controlled or vIGA-AD ≥2
Roflumilast cream 0.05%/0.15% BIW
Endpoints
Primary
• TEAEs
• SAEs
Key secondary
• vIGA-AD
• EASI
• WI-NRS
Other
• Tolerability
• 'Disease control'
Treatment week: 0 4 4 8 16 28 40 56

Application of nonmedicated emollients or moisturizers was allowed for areas where study treatment was not applied.

[a]After OLE study enrollment commenced, the protocol was amended to allow patients aged 2–5 years who completed INTEGUMENT-PED to enroll in INTEGUMENT-OLE.

RESULTS

- Characteristics at parent study baseline were well balanced for the 562 patients (from roflumilast cream 0.05%, n=382; from vehicle cream, n=180) who rolled over into the INTEGUMENT-OLE trial
- Roflumilast cream 0.05%/0.15% was well tolerated
 - Treatment-related AEs were reported for 2.5% of patients; all were mild or moderate
 - Application-site pain was reported as an AE for 4 (0.7%) patients
 - Investigators reported similar application-site tolerability (>98% no/minimal irritation) for patients who received roflumilast cream 0.05% or vehicle cream in the parent trial at all time points; no application-site irritation was reported for ≥97% of patients by investigators throughout the OLE
- Improvements in signs and symptoms of AD observed at the end of INTEGUMENT-PED were maintained or improved further over 52 weeks in patients who continued with roflumilast cream treatment and those who switched from vehicle to roflumilast cream
 - 54.2% of patients overall achieved vIGA-AD success
 - 63.1% achieved vIGA-AD 0/1
 - 73.3% achieved EASI-75
 - 60.7% achieved WI-NRS success
- There were 55 patients who aged to 6 years during either the INTEGUMENT-PED or INTEGUMENT-OLE study and applied at least one dose of roflumilast cream 0.15%
- There were 170 (30.2%) patients who switched to BIW application
 - These patients experienced K-M median duration of 'disease control' of 238 days (34 weeks)

Patient Demographics and Baseline Disease Characteristics[a]

Characteristic		Roflumilast cream 0.05%/0.15% (n=382)	Vehicle to Roflumilast cream 0.05%/0.15% (n=180)	Overall (n=562)
Age, years, mean (SD) [range]		3.3 (1.1) [2–5]	3.2 (1.1) [2–5]	3.3 (1.1) [2–5]
Male at birth, n (%)		197 (51.6)	89 (49.4)	286 (50.9)
Not Hispanic or Latino, n (%)		310 (81.2)	155 (86.1)	465 (82.7)
Race, n (%)	White	262 (68.6)	137 (76.1)	399 (71.0)
	Black or African-American	58 (15.2)	22 (12.2)	80 (14.2)
	Asian	30 (7.9)	15 (8.3)	45 (8.0)
	Multiple	25 (6.5)	3 (1.7)	28 (5.0)
	Other	7 (1.8)	3 (1.7)	10 (1.8)
Fitzpatrick skin type, n (%)[b]	Type I–III	248 (64.9)	126 (70.0)	374 (66.5)
	Type IV–VI	134 (35.1)	53 (29.4)	187 (33.3)
vIGA-AD, n (%)	2 (mild)	86 (22.5)	35 (19.4)	121 (21.5)
	3 (moderate)	296 (77.5)	145 (80.6)	441 (78.5)
Mean (median) [range]	EASI	12.4 (10.6) [4.6–42.0]	11.6 (9.7) [5.0–32.9]	12.2 (10.2) [4.6–42.0]
	BSA, %	22.9 (17.8) [3.0–82.0]	20.9 (17.0) [4.0–75.8]	22.3 (17.5) [3.0–82.0]
	WI-NRS	6.2 (6.0) [0–10]	5.8 (6.0) [0–10]	6.1 (6.0) [0–10]
Prior inadequate response, intolerance, and/or contraindication to TCS		201 (52.6)	96 (53.3)	297 (52.8)

Safety population. [a]Baseline of the parent study (INTEGUMENT-PED). [b]There was one missing value in the vehicle group for Fitzpatrick skin type.

Summary of AEs

Patients, n (%)		Roflumilast cream 0.05%/0.15% (n=562)
Patients with any TEAE		280 (49.8)
Patients with any treatment-related AE		14 (2.5)
Patients with any SAE[a]		18 (3.2)
Patients with any treatment-related SAE		0
Patients who discontinued roflumilast [trial] because of an AE		18 (3.2) [17 (3.0)]
Most common TEAEs by preferred term, ≥4% of patients overall	Upper respiratory tract infection	49 (8.7)
	Nasopharyngitis	28 (5.0)
	Pyrexia	28 (5.0)
	Influenza	22 (3.9)

Safety population. [a]SAEs were pneumonia (n=3), bronchial hyperreactivity (n=2), bronchitis (n=2), and 1 each of abscess jaw, anaphylactic shock, application-site cellulitis, dyspnea, eczema infected, gastroenteritis, gastrointestinal bacterial overgrowth, hypoglycemia, hypoxia, influenza, joint swelling, laryngitis, lymphadenopathy, penetrating abdominal trauma, pneumonia viral, respiratory syncytial virus infection, skin bacterial infection, viral upper respiratory tract infection, and vomiting. Two patients reported three SAEs and four patients reported two SAEs.

Durable Efficacy Responses Over 52 Weeks

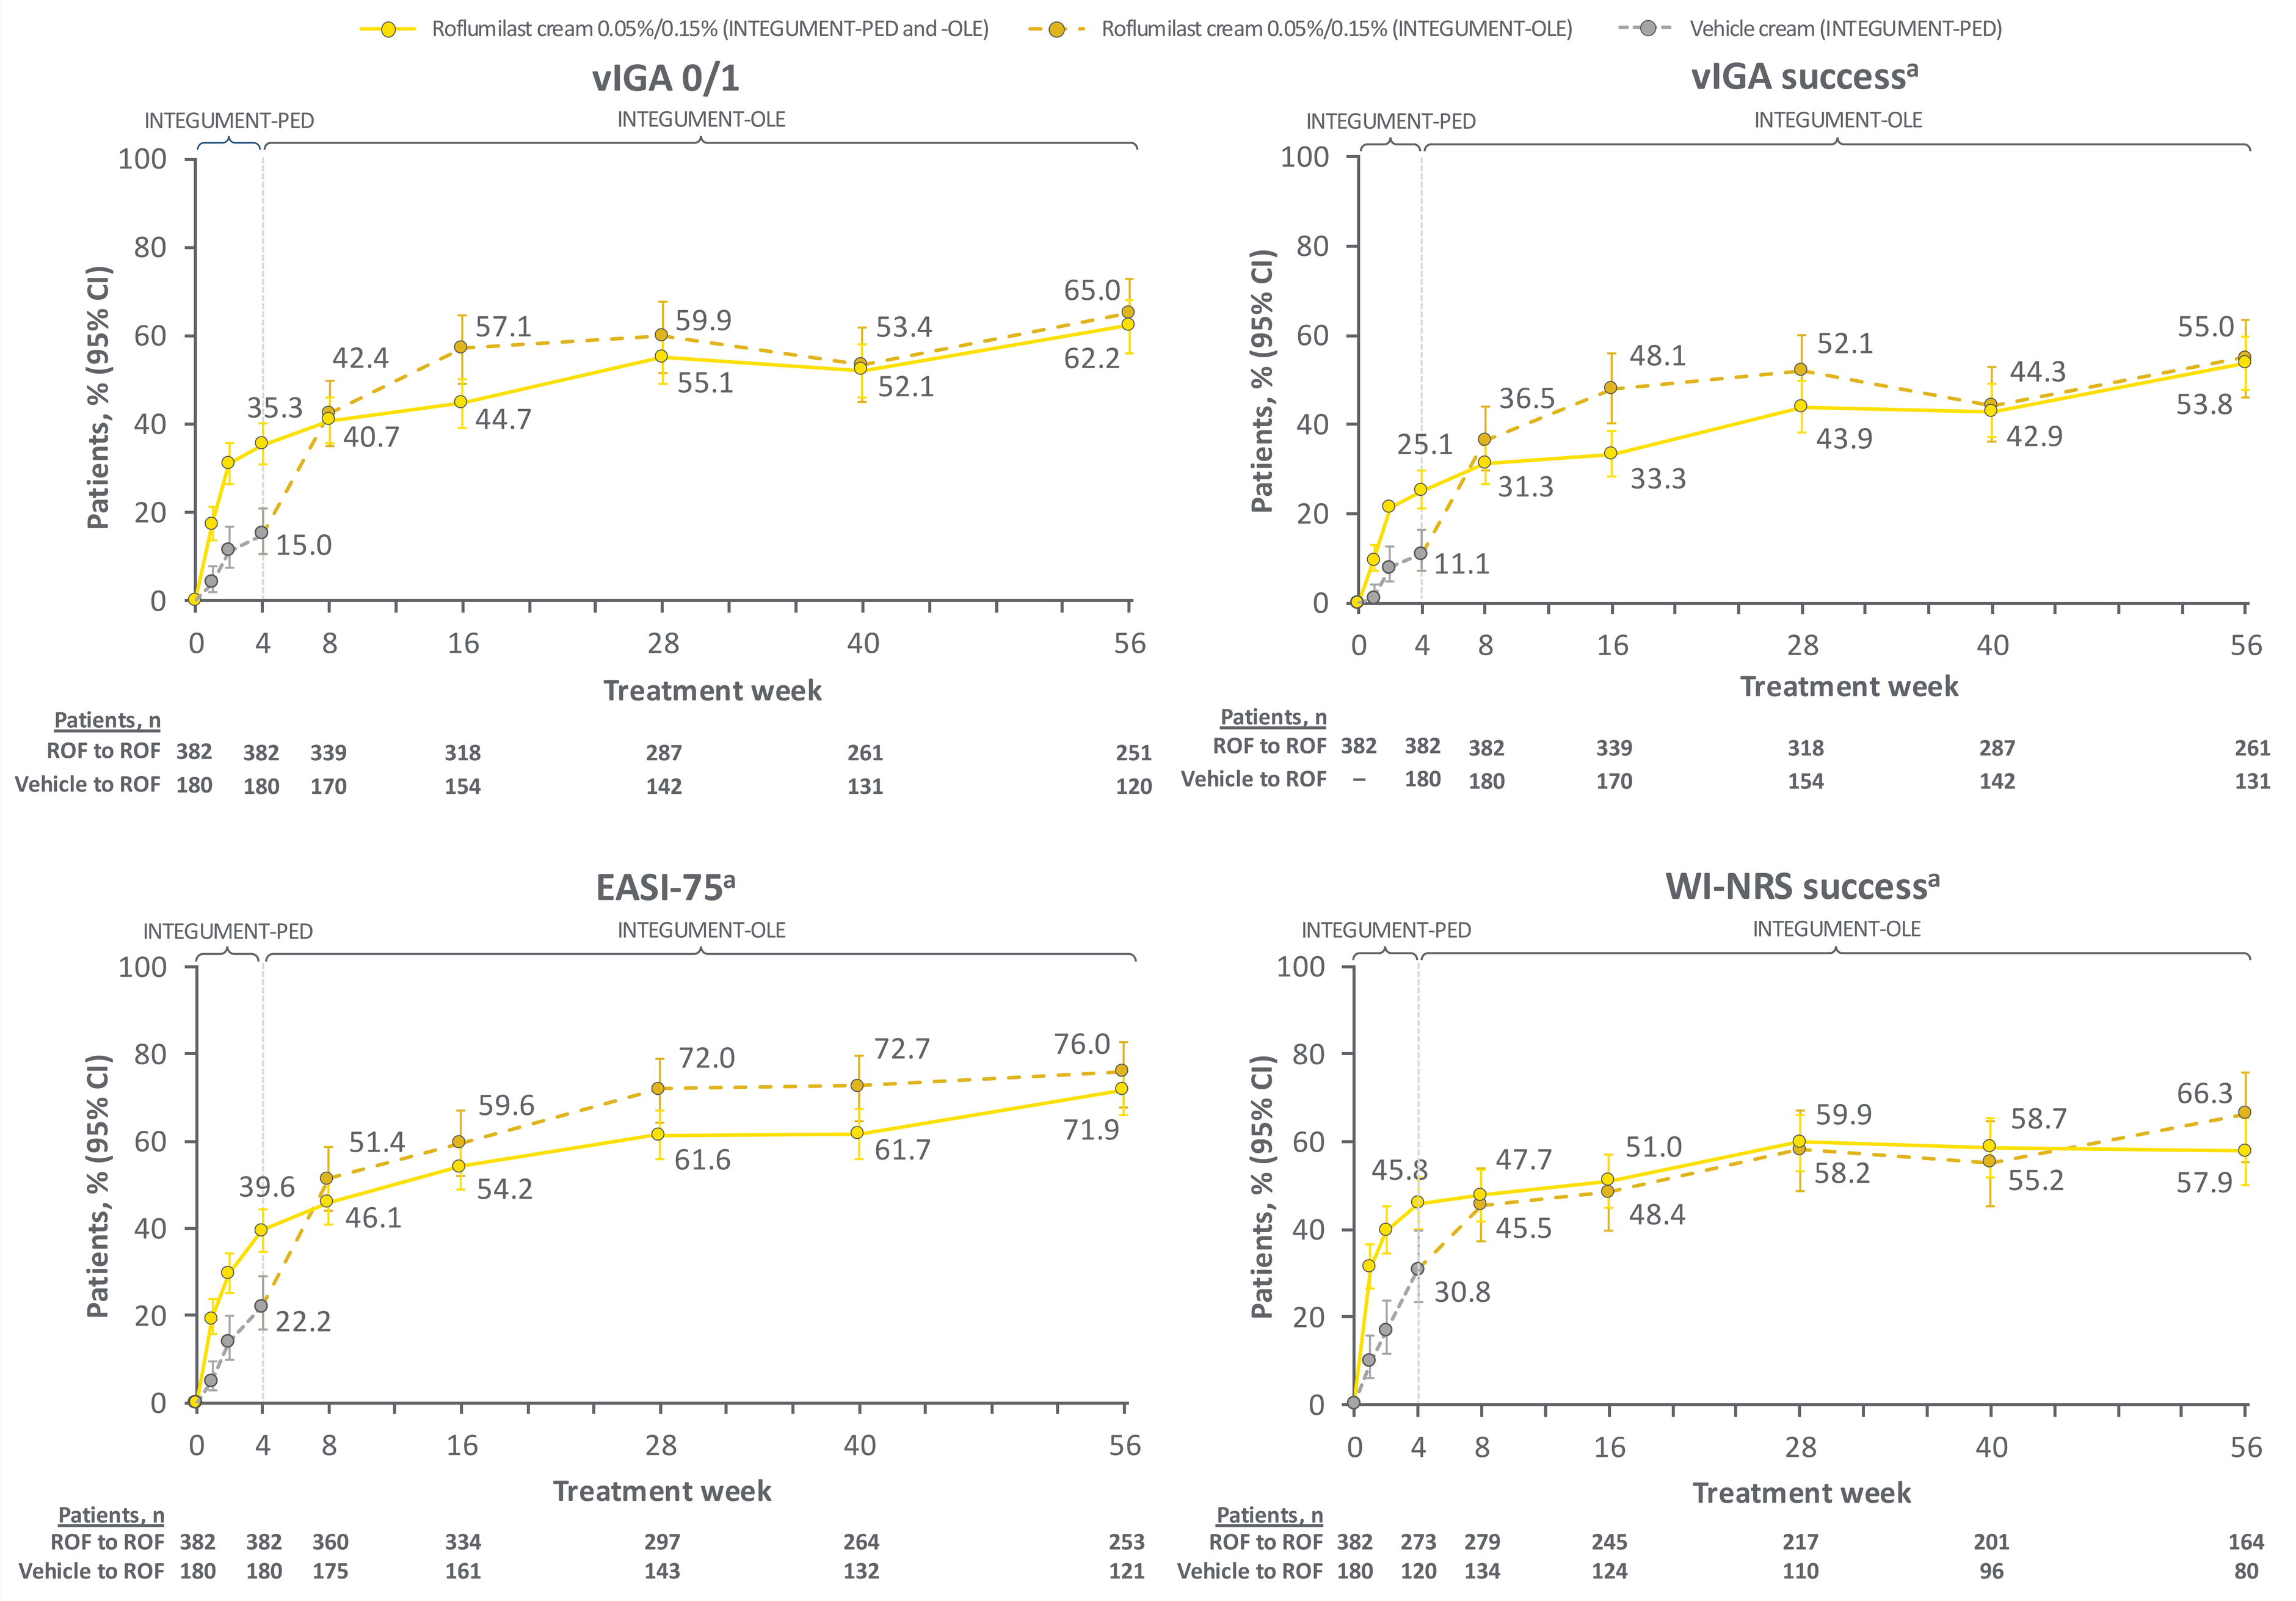

Roflumilast cream 0.05%/0.15% (INTEGUMENT-PED and -OLE)
Roflumilast cream 0.05%/0.15% (INTEGUMENT-OLE)
Vehicle cream (INTEGUMENT-PED)
vIGA 0/1
INTEGUMENT-PED
INTEGUMENT-OLE
Patients, % (95% CI)
35.3 42.4 40.7 15.0 57.1 44.7 59.9 55.1 53.4 52.1 65.0 62.2
Treatment week
0 4 8 16 28 40 56
Patients, n
ROF to ROF 382 382 339 318 287 261 251
Vehicle to ROF 180 180 170 154 142 131 120
vIGA success[a]
25.1 36.5 31.3 11.1 48.1 33.3 52.1 43.9 44.3 42.9 55.0 53.8
Patients, n
ROF to ROF 382 382 382 339 318 287 261
Vehicle to ROF – 180 180 170 154 142 131
EASI-75[a]
39.6 51.4 46.1 22.2 59.6 54.2 72.0 61.6 72.7 61.7 76.0 71.9
Patients, n
ROF to ROF 382 382 360 334 297 264 253
Vehicle to ROF 180 180 175 161 143 132 121
WI-NRS success[a]
45.8 47.7 45.5 30.8 51.0 48.4 59.9 58.2 58.7 55.2 66.3 57.9
Patients, n
ROF to ROF 382 273 279 245 217 201 164
Vehicle to ROF 180 120 134 124 110 96 80

FAS population; observed data. [a]Based on parent study baseline.

Application-Site Tolerability

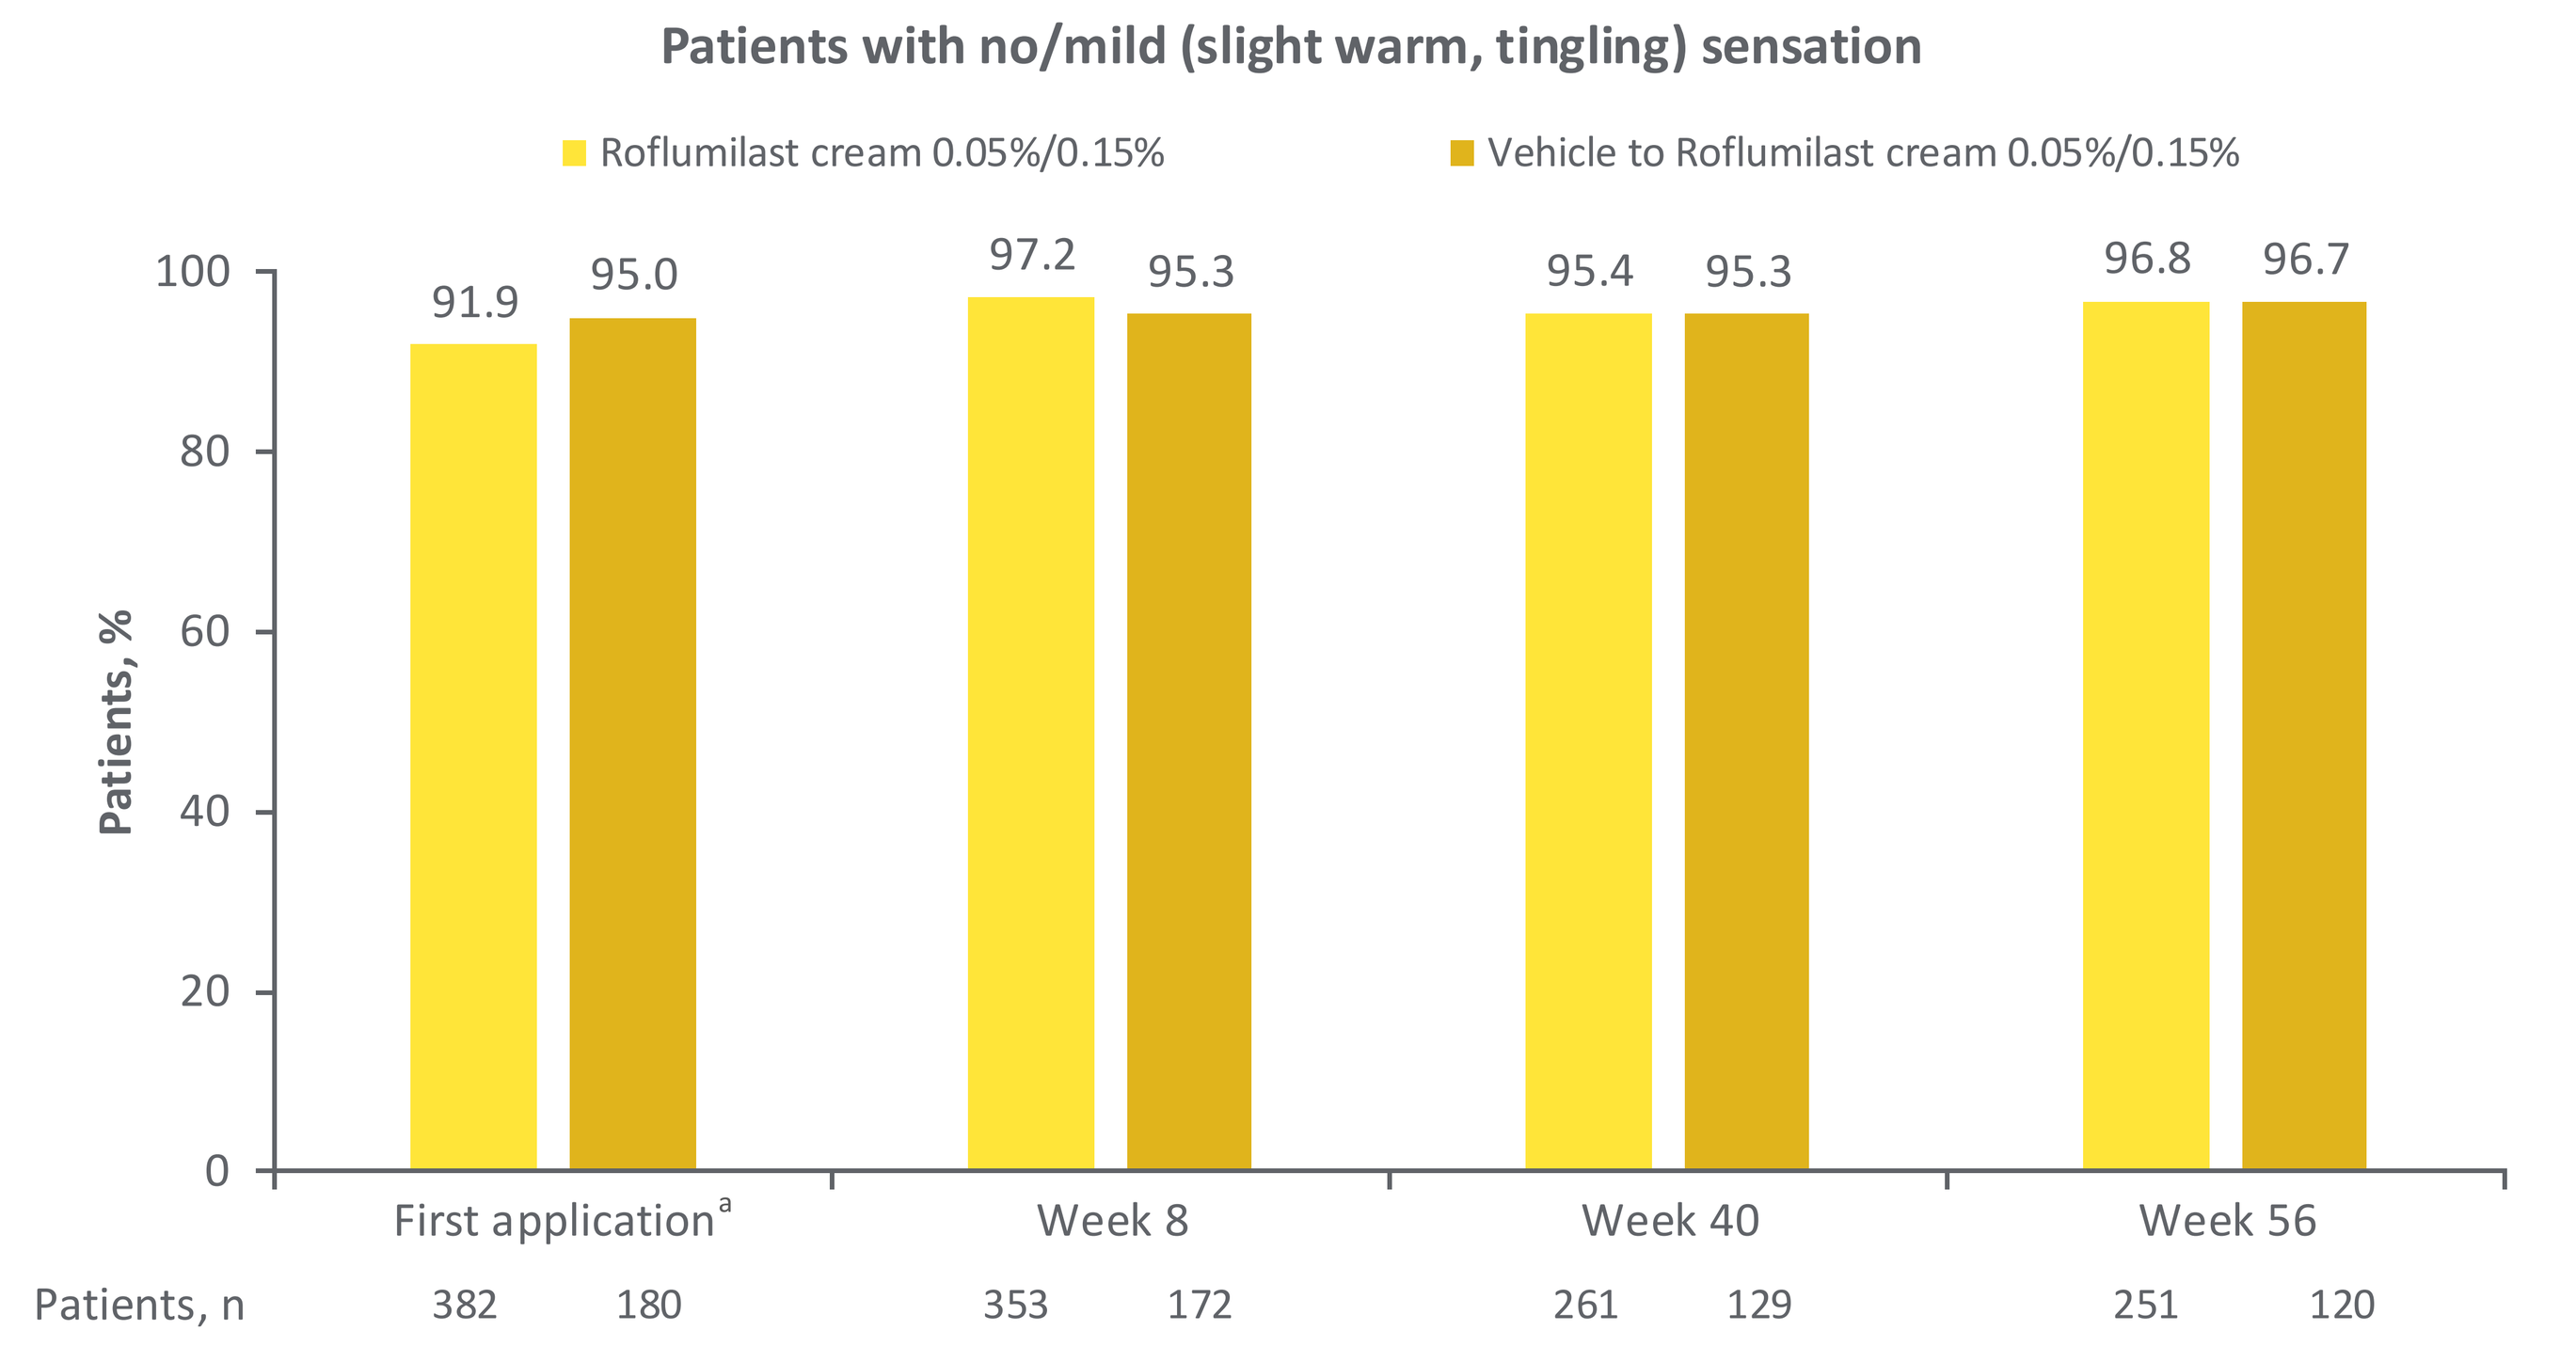

Patients with no/mild (slight warm, tingling) sensation
Roflumilast cream 0.05%/0.15%
Vehicle to Roflumilast cream 0.05%/0.15%
Patients, %
91.9 95.0 97.2 95.3 95.4 95.3 96.8 96.7
First application[a] Week 8 Week 40 Week 56
Patients, n 382 180 353 172 261 129 251 120

[a]First application assessment is 10–15 minutes after the initial application of roflumilast in the parent study or the initial application of roflumilast in INTEGUMENT-OLE for those who received vehicle in the parent study.

Improved Signs and Symptoms of AD Over Time

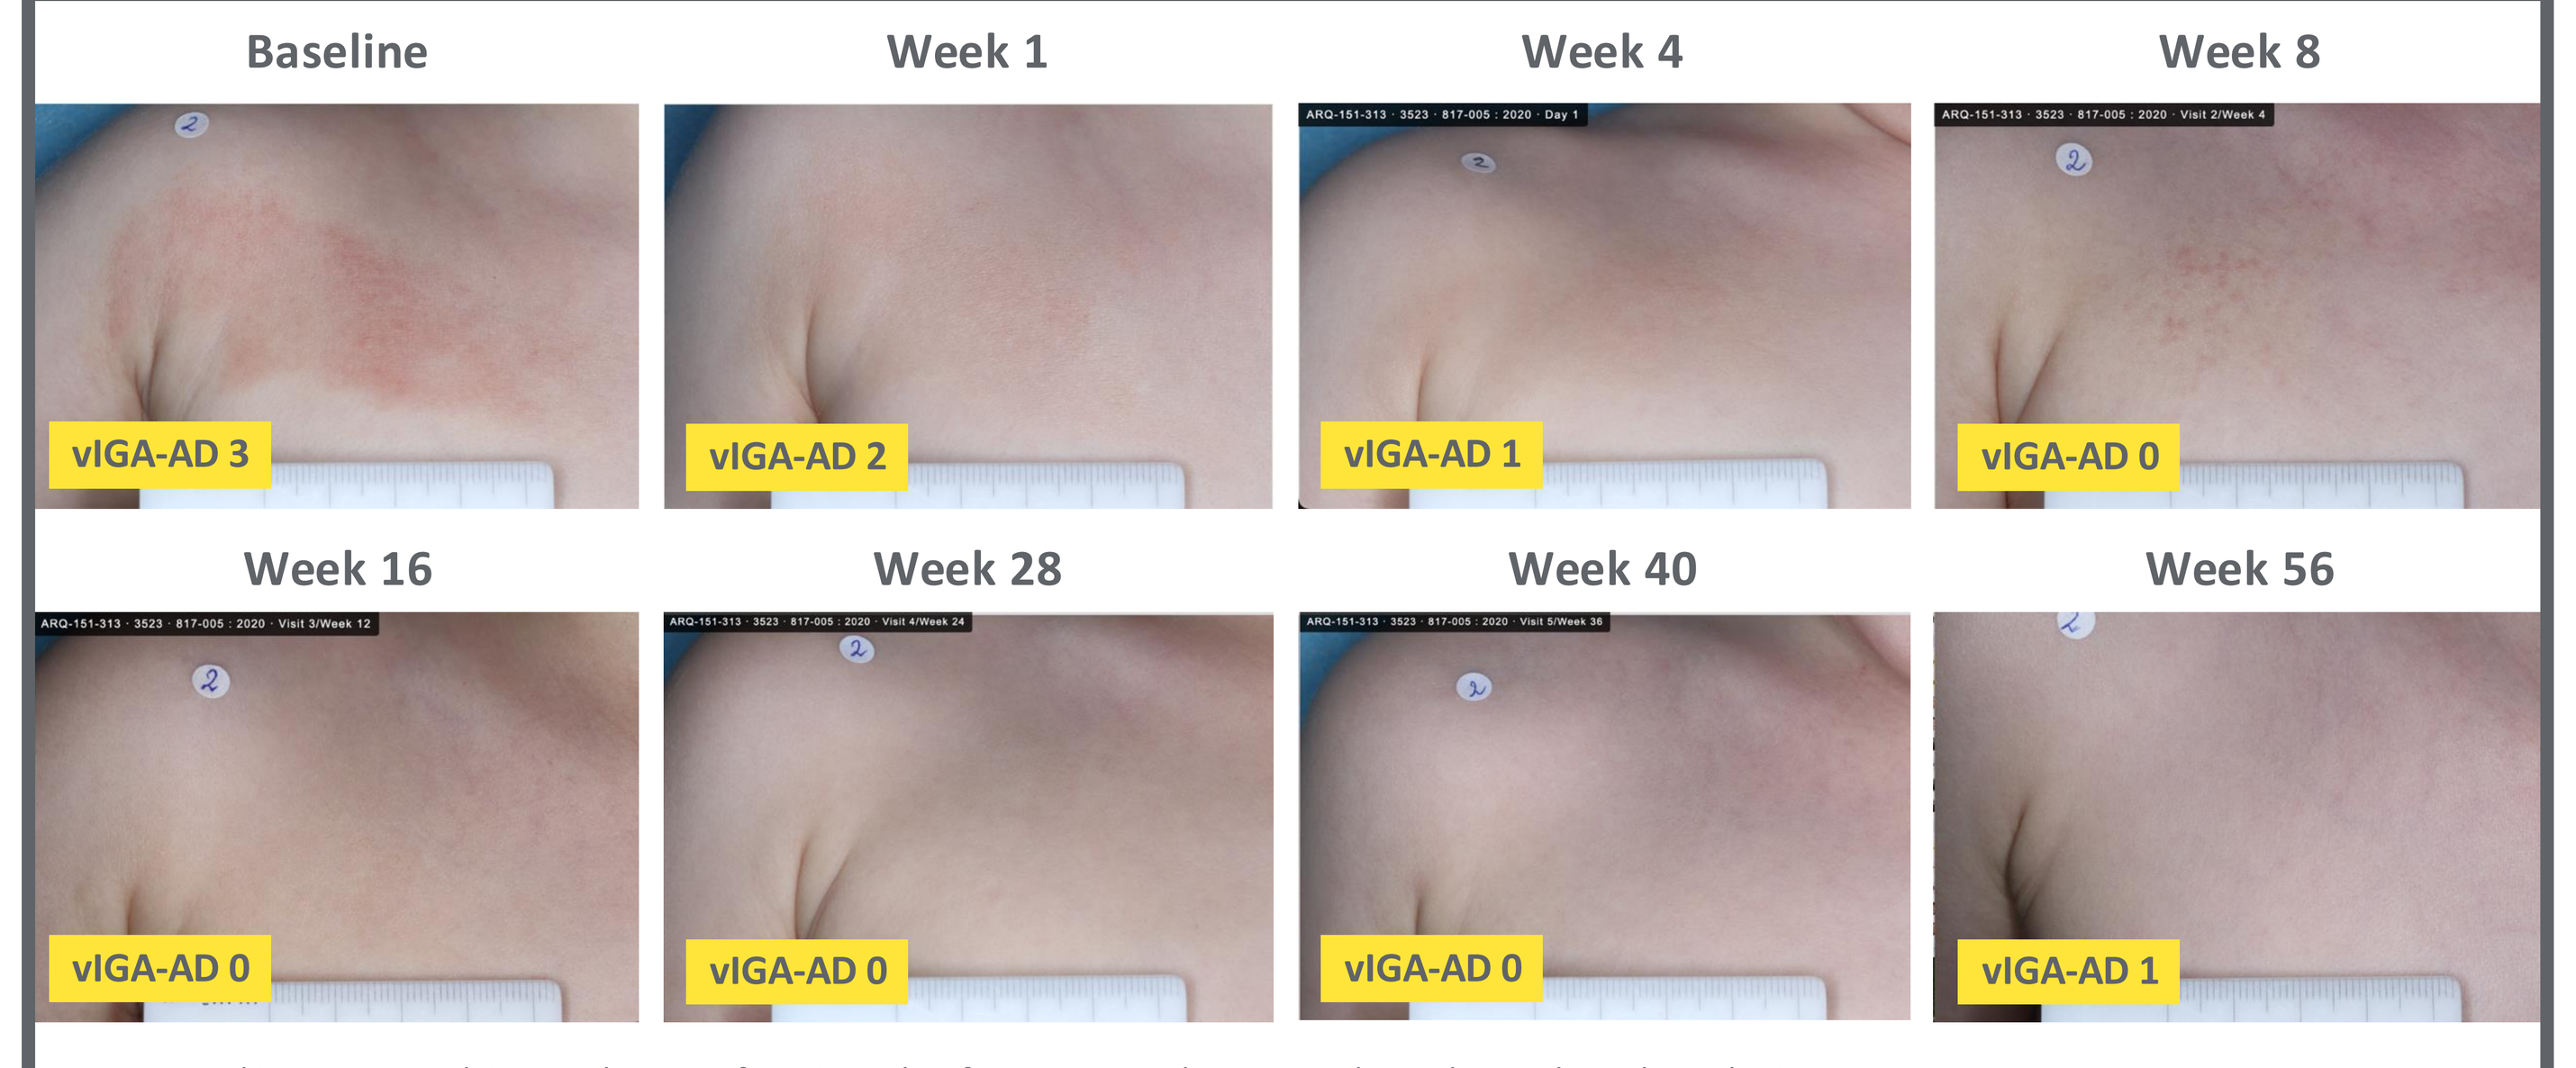

Baseline
Week 1
Week 4
Week 8
vIGA-AD 3
vIGA-AD 2
vIGA-AD 1
vIGA-AD 0
Week 16
Week 28
Week 40
Week 56
vIGA-AD 0
vIGA-AD 0
vIGA-AD 0
vIGA-AD 1

Patient aged 2 years started BIW application after 16 weeks of treatment and maintained BIW dosing throughout the OLE.

'Disease Control' With BIW Application

30.2% of patients achieved vIGA-AD 0 and **switched to proactive BIW application**

Median duration of 'disease control'[a] with BIW application **238 DAYS** (95 CI%, 146–331)

FAS population. [a]K-M estimate.

CONCLUSIONS

- Roflumilast cream 0.05%/0.15% was well tolerated in children aged 2–5 years with AD, for up to 56 weeks
 - There were few SAEs and none were considered related to study treatment
- Durable efficacy was demonstrated for up to 56 weeks of treatment with roflumilast cream 0.05%/0.15%
 - Improvements in AD after 4 weeks of treatment in the parent study improved or were maintained
- Signs and symptoms of AD were proactively managed with BIW application in a subset of patients who achieved vIGA-AD 0, with a K-M median duration of 'disease control' of 238 days
- Safety and efficacy were consistent with results in adults and pediatric patients aged ≥6 years[7,8]
- These results suggest that roflumilast cream is a long-term treatment option for those with AD, with the potential to switch to twice-weekly proactive application